Under-reporting of Symptoms in Patients with Early Breast Cancer who have Received Tamoxifen Treatment for 2 to 3 years

R C Coombes†, J M Bliss‡, E Hall‡, L J Gibson†, L Fallowfield⸸, M Price⸸, and G Massimini¥ on behalf of the IES Steering Committee

†Imperial College Faculty of Medicine, Charing Cross Hospital, London, UK ‡Clinical Trials & Statistics Unit, Institute of Cancer Research, Sutton, UK ⸸Cancer Research UK Psychosocial Oncology Group, University of Sussex, UK ¥Pfizer Clinical Development, Viale Pasteur 10, Nerviano, Italy

ABSTRACT

The Intergroup Exemestane Study (96OEXE031-C/13/96 – BIG02/97) is a randomised double-blind trial assessing the benefit of sequential exemestane after 2-3 years tamoxifen versus tamoxifen alone for a total of 5 years endocrine treatment. Baseline data are available on 4439 postmenopausal patients (mean age 64; range 32-96) with primary breast cancer (48.1% node-positive, of whom 32.7% had received adjuvant chemotherapy and 78.5% known to have ER positive tumours). Hormone therapy is viewed by many as less toxic than many other cancer treatments and side- effects described as minimal or well-tolerated[1]. There is some evidence that menopausal symptoms reach a peak by 12 weeks of treatment and plateau out thereafter[2]. The Intergroup Exemestane Study provides an opportunity to examine the patient-reported side effects of women who have been taking tamoxifen for 2 to 3 years. We have examined (a) baseline symptoms prior to randomisation and (b) the comparative reporting of symptoms from clinician-completed CRFs and patient-reported QOL questionnaires for women participating in the sub-protocol evaluating Quality of Life (QOL). QOL is assessed using the FACT-ES questionnaire[2]. Salient clinical symptoms (all at CTC grade 3) at entry included: headaches 0.5%; hot flushes 3.6%; sweating 0.5%; insomnia 0.4%. Any incidence of thromboembolic disease was reported in 0.2% patients. We have cross-tabulated investigator-reported clinical symptoms by patient-reported symptoms. This showed that some symptoms were recorded similarly using the two methods. These were (as % concordance): vomiting 98%; vaginal bleeding 96.9%; nausea 90.1%. Some were only moderately similar: hot flushes 72.6%; dizziness 72.1%; headaches 65.9%; night sweats 54.7%. However, two symptoms had a <50% concordance: lack of energy 44.4% and insomnia 45.5%. The majority of cases of discordance are probably due to under-reporting by those clinicians completing the CRF. Thus % patients complaining of moderate or severe symptoms where the 'none' box was ticked in the CRF were: nausea 2%; vomiting 1%; headaches 11%;hot flushes 9%; dizziness 8%; insomnia 26%; lack of energy 26%; cold sweats 13% ;night sweats 31%. We conclude that there is a discrepancy between clinician-reported and self-reported tamoxifen-related symptoms.

INTRODUCTION

- The Intergroup Exemestane Study (IES) is an international, double-blind, randomised phase III trial in postmenopausal women who have been taking tamoxifen for 2 to 3 years, comparing subsequent tamoxifen to sequential exemestane giving a total of five years endocrine therapy.
- The study has a QOL sub-protocol which includes assessment of endocrine symptoms. Collection of QOL data continues for comparative analysis of patient reported QOL by treatment.
- Endocrine therapies are generally viewed as a gentler, better-tolerated alternative to other breast cancer treatments. However, there is a often a difference in the frequency and severity of some of the side-effects reported by clinicians compared with patient self-report.[3,4]
- 558 patients have been recruited to the QOL sub-protocol from 93 centres in 8 countries: USA (272 patients), United Kingdom (165), Spain (47), Argentina (37), Italy (23), Australia (8), Netherlands (4), New Zealand (2). Baseline characteristics of patients participating in the QOL sub-protocol have been previously reported.[5]
- At trial entry, there is a cohort of women who have been on tamoxifen for between two and three years.
- This analysis examines discrepancies between patient and clinician symptom reporting at baseline.

METHODS

- Symptoms reported at baseline in the patient Case Report Form (CRF) were recorded by CTC grade (Figure 1). Grades 1 & 2 and grades 3 & 4 have been combined. Symptoms have been divided into those explicitly listed on the CRF and those not specifically listed but recorded under 'other'.
- Quality of life was assessed using the FACT-ES questionnaire (items used here are given in figure 2). Patients are asked to endorse specific items 'not at all', 'a little bit', 'somewhat', 'quite a bit' or 'very much'.
- For this analysis 'a little bit' and 'somewhat' have been combined and compared to CTC grade 1 & 2; 'quite a bit' and 'very much' have been combined and are compared to CTC grades 3 & 4; this gives 3-point ordinal scales for each method of assessment
- Differences in the distribution of severity of symptoms are compared between countries using chi-squared based tests
- Kappa statistics (see box) have been used to measure the degree of agreement between clinician assessed and patient reported symptoms at entry to the trial for symptoms where >10% patients reported 'quite a bit' or 'very much', plus those listed on the CRF.

Figure 1: Extract from IES CRF – signs/symptoms at baseline

✓ Signs/Symptoms	NCIC-CTC Grade 1-4
☐ Nausea (GI NAU)	______
☐ Vomiting (GI VOM)	______
☐ Headaches (NE HEAD)	______
☐ Hot flushes (EN FLA)	______
☐ Vaginal bleeding (code as for GU HEM)	______
☐ Visual disturbances (NE VIS)	______
☐ Dizziness (NE DIZ)	______
☐ Insomnia (NE INS)	______
☐ Fatigue (FL LET)	______
☐ Sweating (FL SWE)	______
☐ Thrombo-embolic disease (CD VEN)	______
☐ Other	______

Figure 2: Symptom specific items and Endocrine subscale of the FACT ES QOL questionnaire

Below is a list of statements that other people with your illness have said are important. By circling one number per line please indicate how true each statement has been for you <u>during the past 7 days. Please not 20 I feel nervous has been omitted</u>

	OTHER SYMPTOMS	not at all	a little bit	some-what	quite a bit	very much
1	I have a lack of energy	0	1	2	3	4
2	I have nausea	0	1	2	3	4
27	I am sleeping well	0	1	2	3	4
	ENDOCRINE SYMPTOMS					
39	I have hot flushes	0	1	2	3	4
40	I have cold sweats	0	1	2	3	4
41	I have night sweats	0	1	2	3	4
42	I have vaginal discharge	0	1	2	3	4
43	I have vaginal itching/irritation	0	1	2	3	4
44	I have vaginal bleeding or spotting	0	1	2	3	4
45	I have vaginal dryness	0	1	2	3	4
46	I have pain or discomfort with intercourse	0	1	2	3	4
47	I have lost interest in sex	0	1	2	3	4
48	I have gained weight	0	1	2	3	4
49	I feel lightheaded/dizzy	0	1	2	3	4
50	I have been vomiting	0	1	2	3	4
51	I have diarrhoea	0	1	2	3	4
52	I get headaches	0	1	2	3	4
53	I feel bloated	0	1	2	3	4
54	I have breast sensitivity/tenderness	0	1	2	3	4
55	I have mood swings	0	1	2	3	4
56	I am irritable	0	1	2	3	4

RESULTS

Table 1: Frequencies of symptoms at entry to the trial

symptoms	CTC grade from CRF	CRF %	self-reported %	QOL score
hot flushes	1 & 2 3 & 4	46.2 3.6	35.3 38.2	a little bit & somewhat quite a bit & very much
weight gain ¥	1 & 2 3 & 4	2.9 0.0	39.1 34.2	a little bit & somewhat quite a bit & very much
sweating (night sweats)	1 & 2 3 & 4	16.3 0.5	29.7 27.1	a little bit & somewhat quite a bit & very much
libido decrease ¥	1 & 2 3 & 4	0.4 0.0	33.7 21.5	a little bit & somewhat quite a bit & very much
insomnia	1 & 2 3 & 4	17.6 0.4	50.2 19.2	a little bit & somewhat quite a bit & very much
vaginal dryness ¥	1 & 2 3 & 4	2.0 0.2	30.8 15.8	a little bit & somewhat quite a bit & very much
headaches	1 & 2 3 & 4	15.2 0.5	34.6 14.2	a little bit & somewhat quite a bit & very much
fatigue	1 & 2 3 & 4	20.3 0.7	57.7 13.8	a little bit & somewhat quite a bit & very much
short of breath ¥	1 & 2 3 & 4	1.1 0.0	31.7 13.4	a little bit & somewhat quite a bit & very much
dizziness	1 & 2 3 & 4	9.5 0.0	28.1 3.9	a little bit & somewhat quite a bit & very much
vaginal bleeding	1 & 2 3 & 4	2.7 0.0	4.1 1.3	a little bit & somewhat quite a bit & very much

Symptoms with '¥' are not specifically listed in the CRF but were recorded under 'other'

Table 2: Frequencies of QOL symptoms not recorded on the CRF

Symptom	Self reported %	QOL Score	Symptom	Self reported %	QOL Score
bloating	36.0 13.8	a little bit & somewhat quite a bit & very much	vaginal discharge	36.8 7.7	a little bit & somewhat quite a bit & very much
breast tenderness	33.3 12.7	a little bit & somewhat quite a bit & very much	vaginal itching	24.8 6.0	a little bit & somewhat quite a bit & very much
mood swings	48.3 11.0	a little bit & somewhat quite a bit & very much	irritable	50.6 5.2	a little bit & somewhat quite a bit & very much
cold sweats	17.8 9.9	a little bit & somewhat quite a bit & very much	diarrhoea	9.6 1.8	a little bit & somewhat quite a bit & very much
lymphoedema	28.7 9.5	a little bit & somewhat quite a bit & very much	vomiting	2.0 0	a little bit & somewhat quite a bit & very much
pain with intercourse	24.4 8.7	a little bit & somewhat quite a bit & very much			

There were some differences in the frequency distributions of both clinician and patient reported symptoms by country.

- The distribution of clinical symptom scores differed significantly (p<0.01) by country for hot flushes, sweating, headaches and fatigue.
- Levels of patient reporting differed significantly by country for hot flushes and fatigue and also insomnia, night sweats and vaginal dryness.
- As an example, Figure 3 compares the clinician and patient reports of hot flushes by country – clinicians and patients from the US are more likely to report hot flushes than patients from other countries.

Kappa (K) statistics measure inter-rater agreement - here the degree of agreement between clinician assessed and patient reported symptoms at entry to the trial
- K=1 when agreement is perfect
- K>0.8 indicates good agreement
- K<0.5 indicates poor-fair agreement
- K=0 when agreement is no better than chance
- Weighted K takes into account the degree of disagreement
- Interpretation of kappa depends on number of categories and prevalence of the symptom across categories

Figure 3: Differential reporting of hot flushes

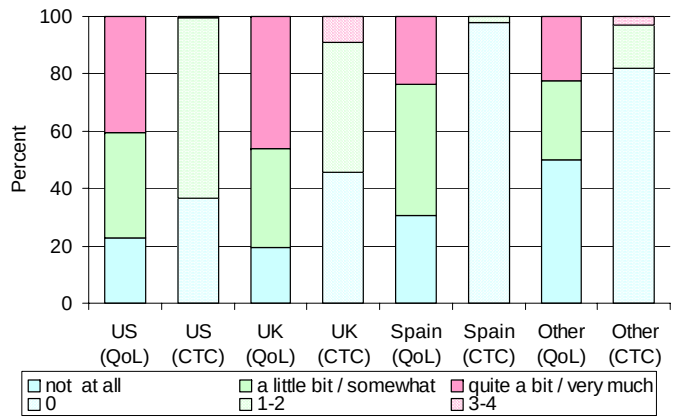

100
80
60
40
20
0
Percent
US (QoL)
US (CTC)
UK (QoL)
UK (CTC)
Spain (QoL)
Spain (CTC)
Other (QoL)
Other (CTC)
not at all
0
a little bit / somewhat
1-2
quite a bit / very much
3-4

Table 3 shows the kappa statistic for scores of none versus any symptoms.

Table 3 Agreement between clinician and patient reporting of presence of symptoms

Symptom	% Prevalence CRF	% Prevalence QOL	Kappa	95% CI
Symptoms listed on CRF				
Hot flushes	49.8	73.5	0.73	0.70 – 0.75
Insomnia	17.9	69.4	0.45	0.42 – 0.48
Headaches	15.8	48.7	0.66	0.63 – 0.69
Fatigue	21.0	71.5	0.44	0.41 – 0.47
Dizziness	9.5	32.1	0.72	0.69 – 0.75
Vaginal bleeding	2.7	5.4	0.97	0.96 – 0.98
Symptoms not explicitly listed on CRF				
Weight gain	2.9	73.3	0.29	0.26 – 0.31
Sweating / night sweats	16.8	56.8	0.55	0.52 – 0.58
Decreased libido	0.4	55.2	0.37	0.34 – 0.40
Vaginal dryness	2.2	46.6	0.54	0.51 – 0.57
Shortness of breath	1.1	45.1	0.62	0.59 - 0.66

Weight gain and decreased libido show only fair agreement (kappa < 0.4) with much higher prevalence being reported directly by patients via the quality of life questionnaires than via their clinician during their baseline assessment.

Table 4 further assesses agreement by considering weighted kappas for 3 point severity scales.

Table 4: Agreement and kappa statistics for scoring of symptoms listed on CRF (3 point scale)

Symptom	Observed agreement	Expected agreement	Weighted kappa	95% CI
Symptoms listed on CRF				
Hot flushes	69%	55%	0.296	0.25 - 0.35
Insomnia	63%	59%	0.098	0.06 - 0.14
Headaches	80%	74%	0.205	0.14 - 0.26
Fatigue	65%	62%	0.097	0.06 - 0.14
Dizziness	83%	80%	0.168	0.11 - 0.23
Vaginal bleeding	97%	95%	0.325	0.25 - 0.39
Symptoms not explicitly listed on CRF				
Weight gain	47%	47%	0.013	0.00 - 0.02
Sweating / night sweats	64%	59%	0.127	0.09 - 0.17
Decreased libido	56%	56%	0.002	0.00 - 0.01
Vaginal dryness	69%	68%	0.035	0.02 - 0.06
Shortness of breath	79%	78%	0.023	0.013-0.051

- As expected, on average, there was significantly less agreement between clinician and patient reported scoring of symptoms for questions that were not explicitly listed on the CRF (mean kappa 0.040 versus 0.198, p=0.01)
- Agreement for 'decreased libido' is no better than that expected by chance.

Table 5 shows weighted kappa statistics (3 point severity scales) by country for the top 3 recruiting countries to the Quality of Life study. Shaded cells indicate agreement is no better than by chance (1% significance level). Country specific frequencies for severe symptoms (CTC score 3 or 4 or patient 'quite a bit' or 'very much') are small thus comparisons are exploratory.

Table 5: Country specific kappa statistics (3 point scale)

Number with baseline CRF	N = 558 All patients Kappa	SE	N = 272 US Kappa	SE	N = 165 UK Kappa	SE	N = 47 Spain Kappa	SE
Symptom listed on CRF								
Hot flushes	**.293**	.023	**.313**	.030	**.278**	.043	**.014**	.021
Insomnia	**.098**	.018	**.116**	.031	**.148**	.038	**.029**	.025
Headaches	**.205**	.030	**.234**	.048	**.282**	.058	**.035**	.034
Fatigue	**.097**	.021	**.054**	.027	**.141**	.039	**.000**	-
Dizziness	**.168**	.029	**.152**	.043	**.267**	.057	**.000**	.000
Vaginal bleeding	**.325**	.034	**.337**	.049	**.265**	.062	**.483**	.128
Symptom not listed on CRF								
Weight gain	**.013**	.006	**.013**	.008	**.017**	.012	Small numbers	
Sweating / night sweats	**.127**	.020	**.124**	.029	**.120**	.035		
Decreased libido	**.002**	.002	**.004**	.005	**.000**	.000		
Vaginal dryness	**.035**	.010	**.038**	.016	**.031**	.016		
Shortness of breath	**.023**	.010	**.047**	.020	**.000**	.000		

- There is no significant difference between average kappa statistics by country
- There are some country differences in degree of agreement on specific symptoms.

CONCLUSIONS

- Clinically severe symptoms (CTC grade 3 or 4) are not commonly reported in this cohort of women who have been on tamoxifen for 2 to 3 years
- The current system of symptom collection, either with open questions or with collection of symptoms by positive list, leaves room for underreporting of symptoms of low clinical severity but high QOL impact.
- Some important symptoms, in particular those to do with sexual issues, are self-reported by a significant proportion of women but are under-reported or not detected at clinical assessment
- Lack of agreement / under-reporting could be due to
 - patients not reporting symptoms to their clinician
 - differences in time period considered
 - clinicians not asking a complete range of questions or open-ended questions
- We are investigating whether the method of CRF completion (e.g. timing of CRF completion relative to patient visit, method of ascertainment of degree of symptoms) further affects reporting of symptoms
- There are some country differences in prevalence of symptoms and degree of agreement on specific symptoms which have implications for QOL studies with wide international participation, particularly for how data are extrapolated to different countries
- The use of specific QOL questionnaires helps in identifying underreported symptoms but careful clinical evaluation and inquiries on other symptoms pertaining to the psychological and social domains should be considered, particularly in low risk patients treated with hormones or other generally well tolerated regimens

References

1. Fellowes D, Fallowfield LJ, Saunders CM, Houghton J (2001) Tolerability of hormone therapies for breast cancer: how informative are documented symptom profiles in medical notes for 'well-tolerated' treatments? Br Cancer Res & Treat 66(1) 73-81
2. Fallowfield LJ, Leaity SK, Howell A, Benson S, Cella D (1999) Assessment of quality of life in women undergoing hormone therapy for rest cancer: validation of an endocrine symptom subscale for the FACT-B Br Cancer Res & Treat 55(2) 189-99
3. Savage C, Pater JL, Tu D,Norris B (2002) He said/she said: how much agreement is there on symptoms between common toxicity criteria and quality of life? Proc ASCO 21 40a 1540
4. Fallowfield L, Cella D on behalf of the ATAC Trialists' Group (2002) Assessing the quality of life (QOL) of postmenopausal (PM) women randomized into the ATAC ('Arimidex', tamoxifen, alone or in combination) adjuvant breast cancer (BC) trial. Proc ASCO 21 40a 159
5. Fallowfield L, Murray RA, Coster S, Coombes RC, Gibson L, Bliss J, Hall E on behalf of the IES Steering Committee (2002) Patient reported side effects after 2 to 3 years adjuvant tamoxifen. Br Cancer Res & Treat 76 [Suppl 1], S139.

Acknowledgments

Groups participating in the IES QOL sub-protocol

Argentine Breast Cancer group (40 patients), Australian Breast Cancer Trials Group (10 patients), European Organisation for Research and Treatment of Cancer (4 patients), Grupo Espanol de Investigacion del Cancer de Mama (50 patients), Gruppo Oncologicao Italiano di Ricerca sul Cancro (12 patients), International Breast Cancer Study Group (3 patients), International Collaborative Cancer Group (84 patients), Italian Trials in Medical Oncology (9 patients), North West England (14 patients), Yorkshire Breast Group (51 patients), Wales Cancer Trials Network (24 patients), US Oncology (280 patients).

Thank you to Pfizer, to Cancer Research UK and to the staff of the IES Coordinating Data Centre in London.